Complete regression and long-term specific protective immune responses obtained in rodent tumor models after intratumoral treatment with LTX-315

Ø. REKDAL, G. KVALHEIM, PD. LINE, B. ROLSTAD, K. CAMILIO, G. BERGE, J. NESTVOLD, MY. WANG, J. SHI, A. AREFFARD, B. SVEINBJØRNSSON,
Oslo University Hospital, The Norwegian Radium Hospital, University of Oslo, University of Tromsø, Lytix Biopharma AS, NO-9294 Tromsø, Norway.

Lytix Biopharma

Background

Host defense peptides are present in most living species and have a diverse range of functions including direct killing of pathogens and immune-modulating properties. Some host defense peptides are more potent against cancer cells than normal eukaryotic cells due to increased number of anionic membrane components at the surface of the plasma membrane (1 -7).

Several host defense peptides have shown to confer a complete and permanent tumor regression of different types of solid tumors in animal models.

Our structure-activity relationship studies on host defense peptides has culminated in the engineering of small peptides with increased activity towards cancer cells. Ultimately, these efforts have led to the development of a chemically modified 9-mer peptide, LTX-315 (Fig 1).

In addition to a direct killing effect, preliminary results also indicate that LTX-315 has direct effects on dendritic cells (data not shown).

A phase 1 study has been completed with LTX-315 (data under analysis) and outline of a further clinical study is in progress.

LTX-315

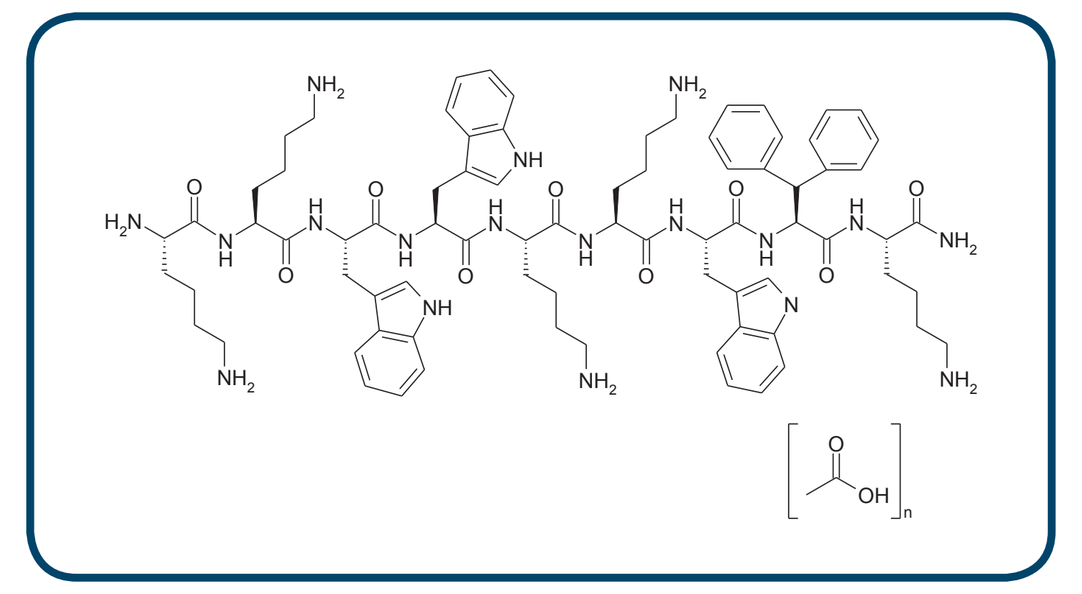

Fig. 1. Chemical structure of LTX-315

Aim

To investigate mode of action and anti-cancer activity by LTX-315 in vitro and in rodent tumor models

Results

- LTX-315 is equally active against drug-sensitive and drug-resistant cancer cells (Table 1)
- In vitro killing kinetic shows that LTX-315 kills cancer cell rapidly compared to conventional chemotherapy (Fig 2)
- Treatment of B16 melanoma cells induced release of HMGB1 (Fig 3)
- LTX-315 induced a complete regression in a rat transformed mesenchymal sarcoma model (rTMSC) (Fig 4)
- Cured animals were protected against re-challenge with TMSC cells s.c. and i.p. (Fig 4)
- Intra-tumoral treatment with LTX-315 leads to early infiltration of granulocytes and at a later stage cytotoxic CD8+ T cells in the rTMSC model (Fig 5)
- An elevated expression of IL1, and IL6 was evident in LTX-315 treated B16F tumor tissue (Fig 6)
- Spleen cells from cured animals prevented tumor growth when transferred into lymphopenic naïve rats (Fig 7)
- Protective immune responses were obtained in a hepatocellular carcinoma (HCC) model when rats were re-challenged with HCC cells, both after complete regression (Fig 8) and after partial responses followed by surgical removal of remaining HCC tumors (Fig 9)
- Complete tumor regression has been obtained in several animal models and protective immune-responses were obtained in all syngeneic models (Table 2)

Table 1: Activity is unaffected by drug-resistance

Cell line	Origin	EC_{50} μM
HL-60	Acute promyelocytic leukemia	2,1
HL-60/ADR	Acute promyelocytic leukemia	3,0
MCF-7	Breast carcinoma	1,9
MCF-7/mdr	Breast carcinoma	2,0
IGROV-1	Ovary carcinoma	6,4
IGROV-1/CDDP	Ovary carcinoma	3,2
K-562	Chronic myeloid leukemia	3,3
K5627/Gleevec	Chronic myeloid leukemia	3,0
HUVEC	Normal endothelial cells	23
	Human Red Blood Cells (normal)	833

Drug-sensitive Drug-resistant Normal cells

- LTX-315 was tested against drug-sensitive and drug-resistant cancer cell pairs from different origins, normal endothelial cells and red blood cells (4h).

Figure 2. In vitro kill kinetic of LTX-315

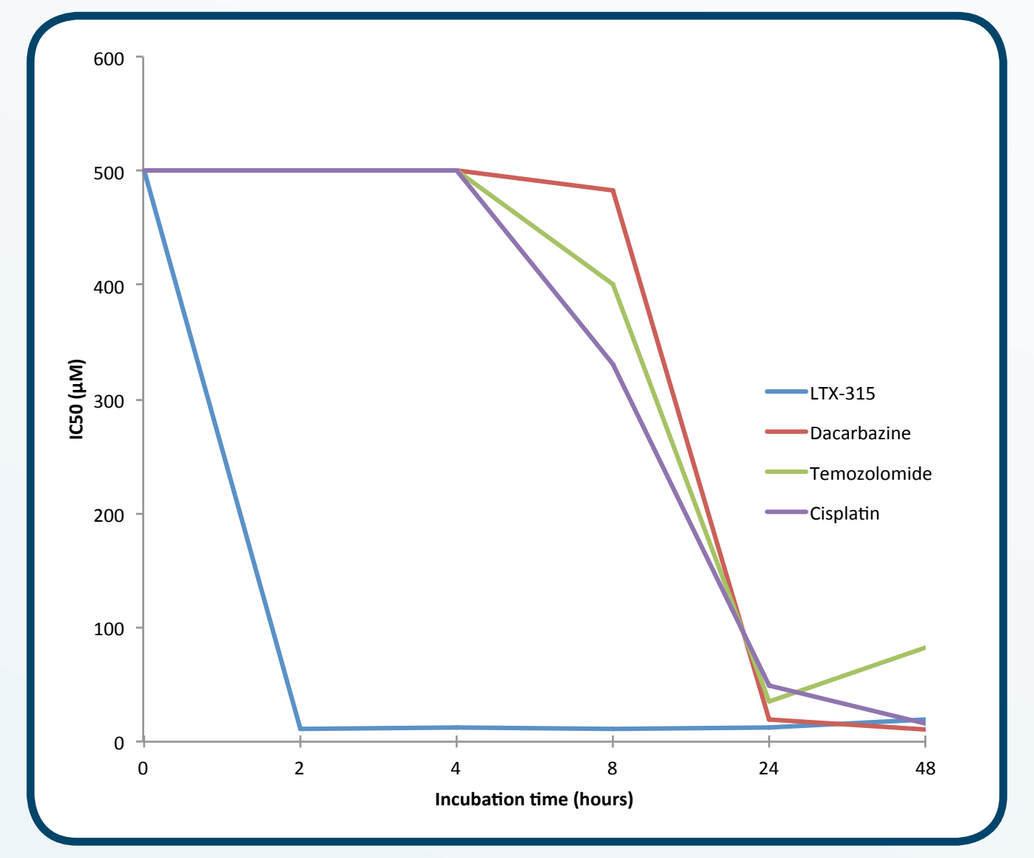
IC50 (μM)
Incubation time (hours)
LTX-315
Dacarbazine
Temozolomide
Cisplatin

- In vitro kill kinetic of LTX-315 against a human A375 melanoma cell line compared with three different chemotherapeutic drugs (dacarbazine, temozolomide and cisplatin).

Figure 3. LTX-315 induced release of the danger signal molecule HMGB1

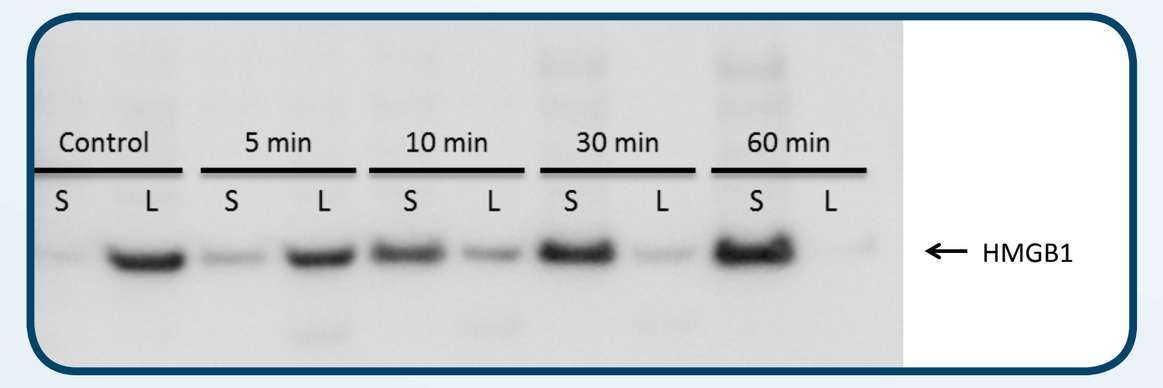
Control
5 min
10 min
30 min
60 min
S L
← HMGB1

HMGB1-release was assessed at different time points by Western blotting of the supernatant (S) or the cell lysate (L) of B16F1 melanoma cells treated with LTX-315 (50mµ/ml).

Figure 4. Effect of LTX-315 in a rat transformed mesenchymal sarcoma model (rTMSC).

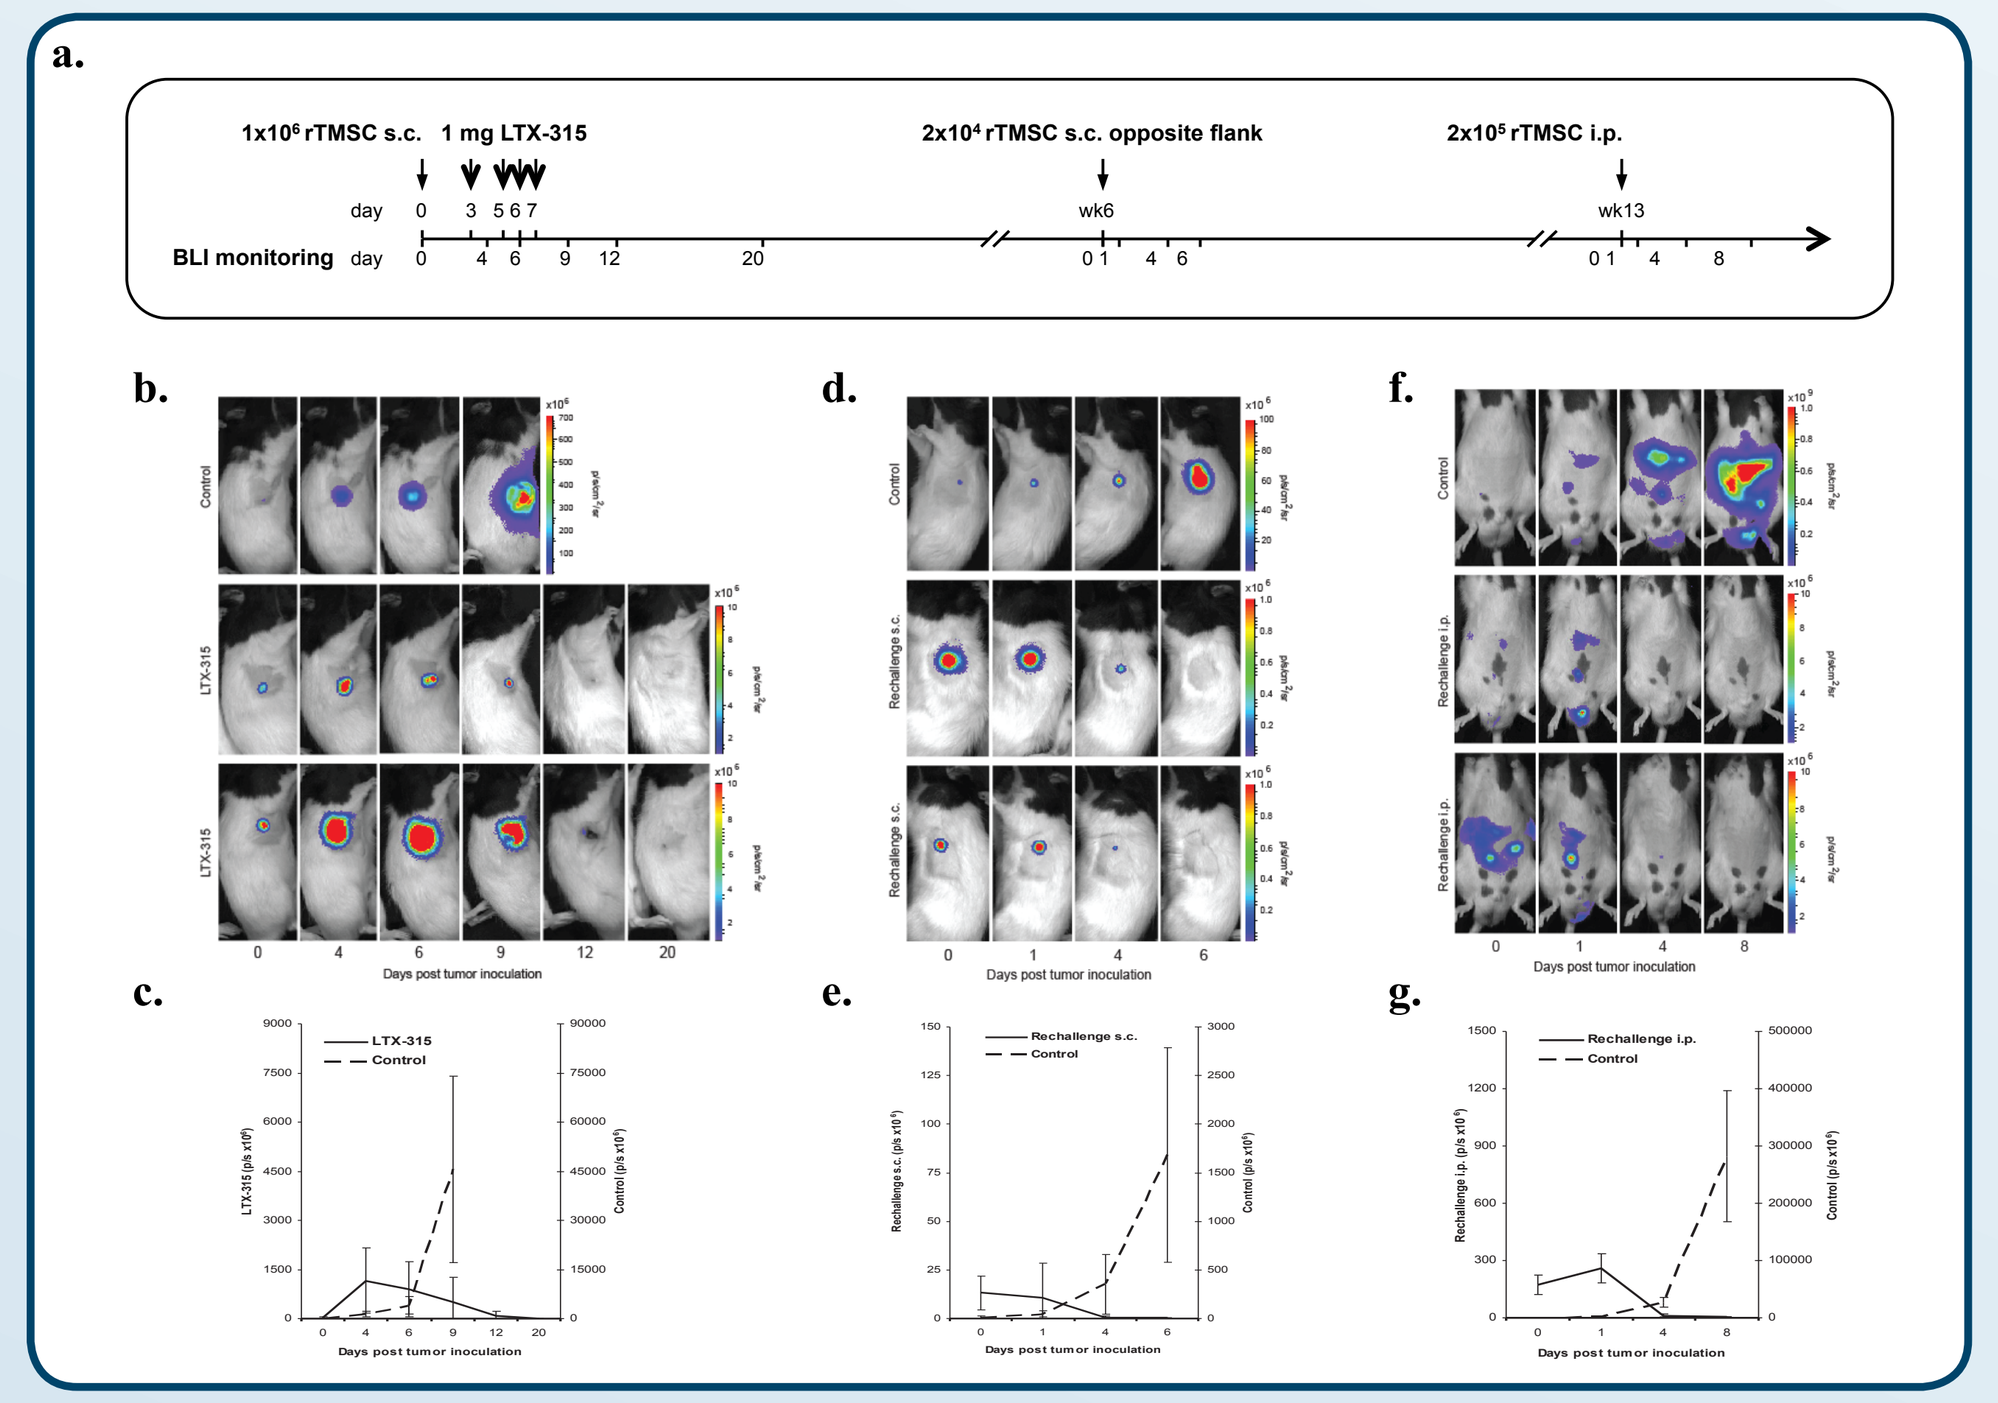
1x10⁶ rTMSC s.c.
1 mg LTX-315
2x10⁴ rTMSC s.c. opposite flank
2x10⁵ rTMSC i.p.
BLI monitoring
Days post tumor inoculation

a. Tumors were established by subcutaneous (s.c.) inoculation of $1{\cdot}10^6$ TMSC in syngeneic PVG rats. At days 3, 5, 6 and 7, the rats (n=6) were treated intra-tumorally (i.t.) with 1 mg LTX-315 (in 50 µL saline).
b. Representative whole-body bioluminescence images of rats during treatment with LTX-315 or vehicle.

c. Total luciferase activity is presented and shows tumor progression and response to LTX-315 treatment compared to control rats (n=6)

d. Representative luminescence images of previously treated animals and controls after s.c. re-challenge with $2{\cdot}10^4$ TMSC

e. Tumor growth in re-challenged and control animals (s.c.) (n=6).

f. Representative luminescence images of animals after intra-peritoneal (i.p) inoculation of $2{\cdot}10^5$ TMSC at week 13.

g. Tumor growth i.p. in re-challenged and control rats. The data are mean ± SD (n=6)

Figure 5. Infiltration of immune cells in LTX-315-treated tumors

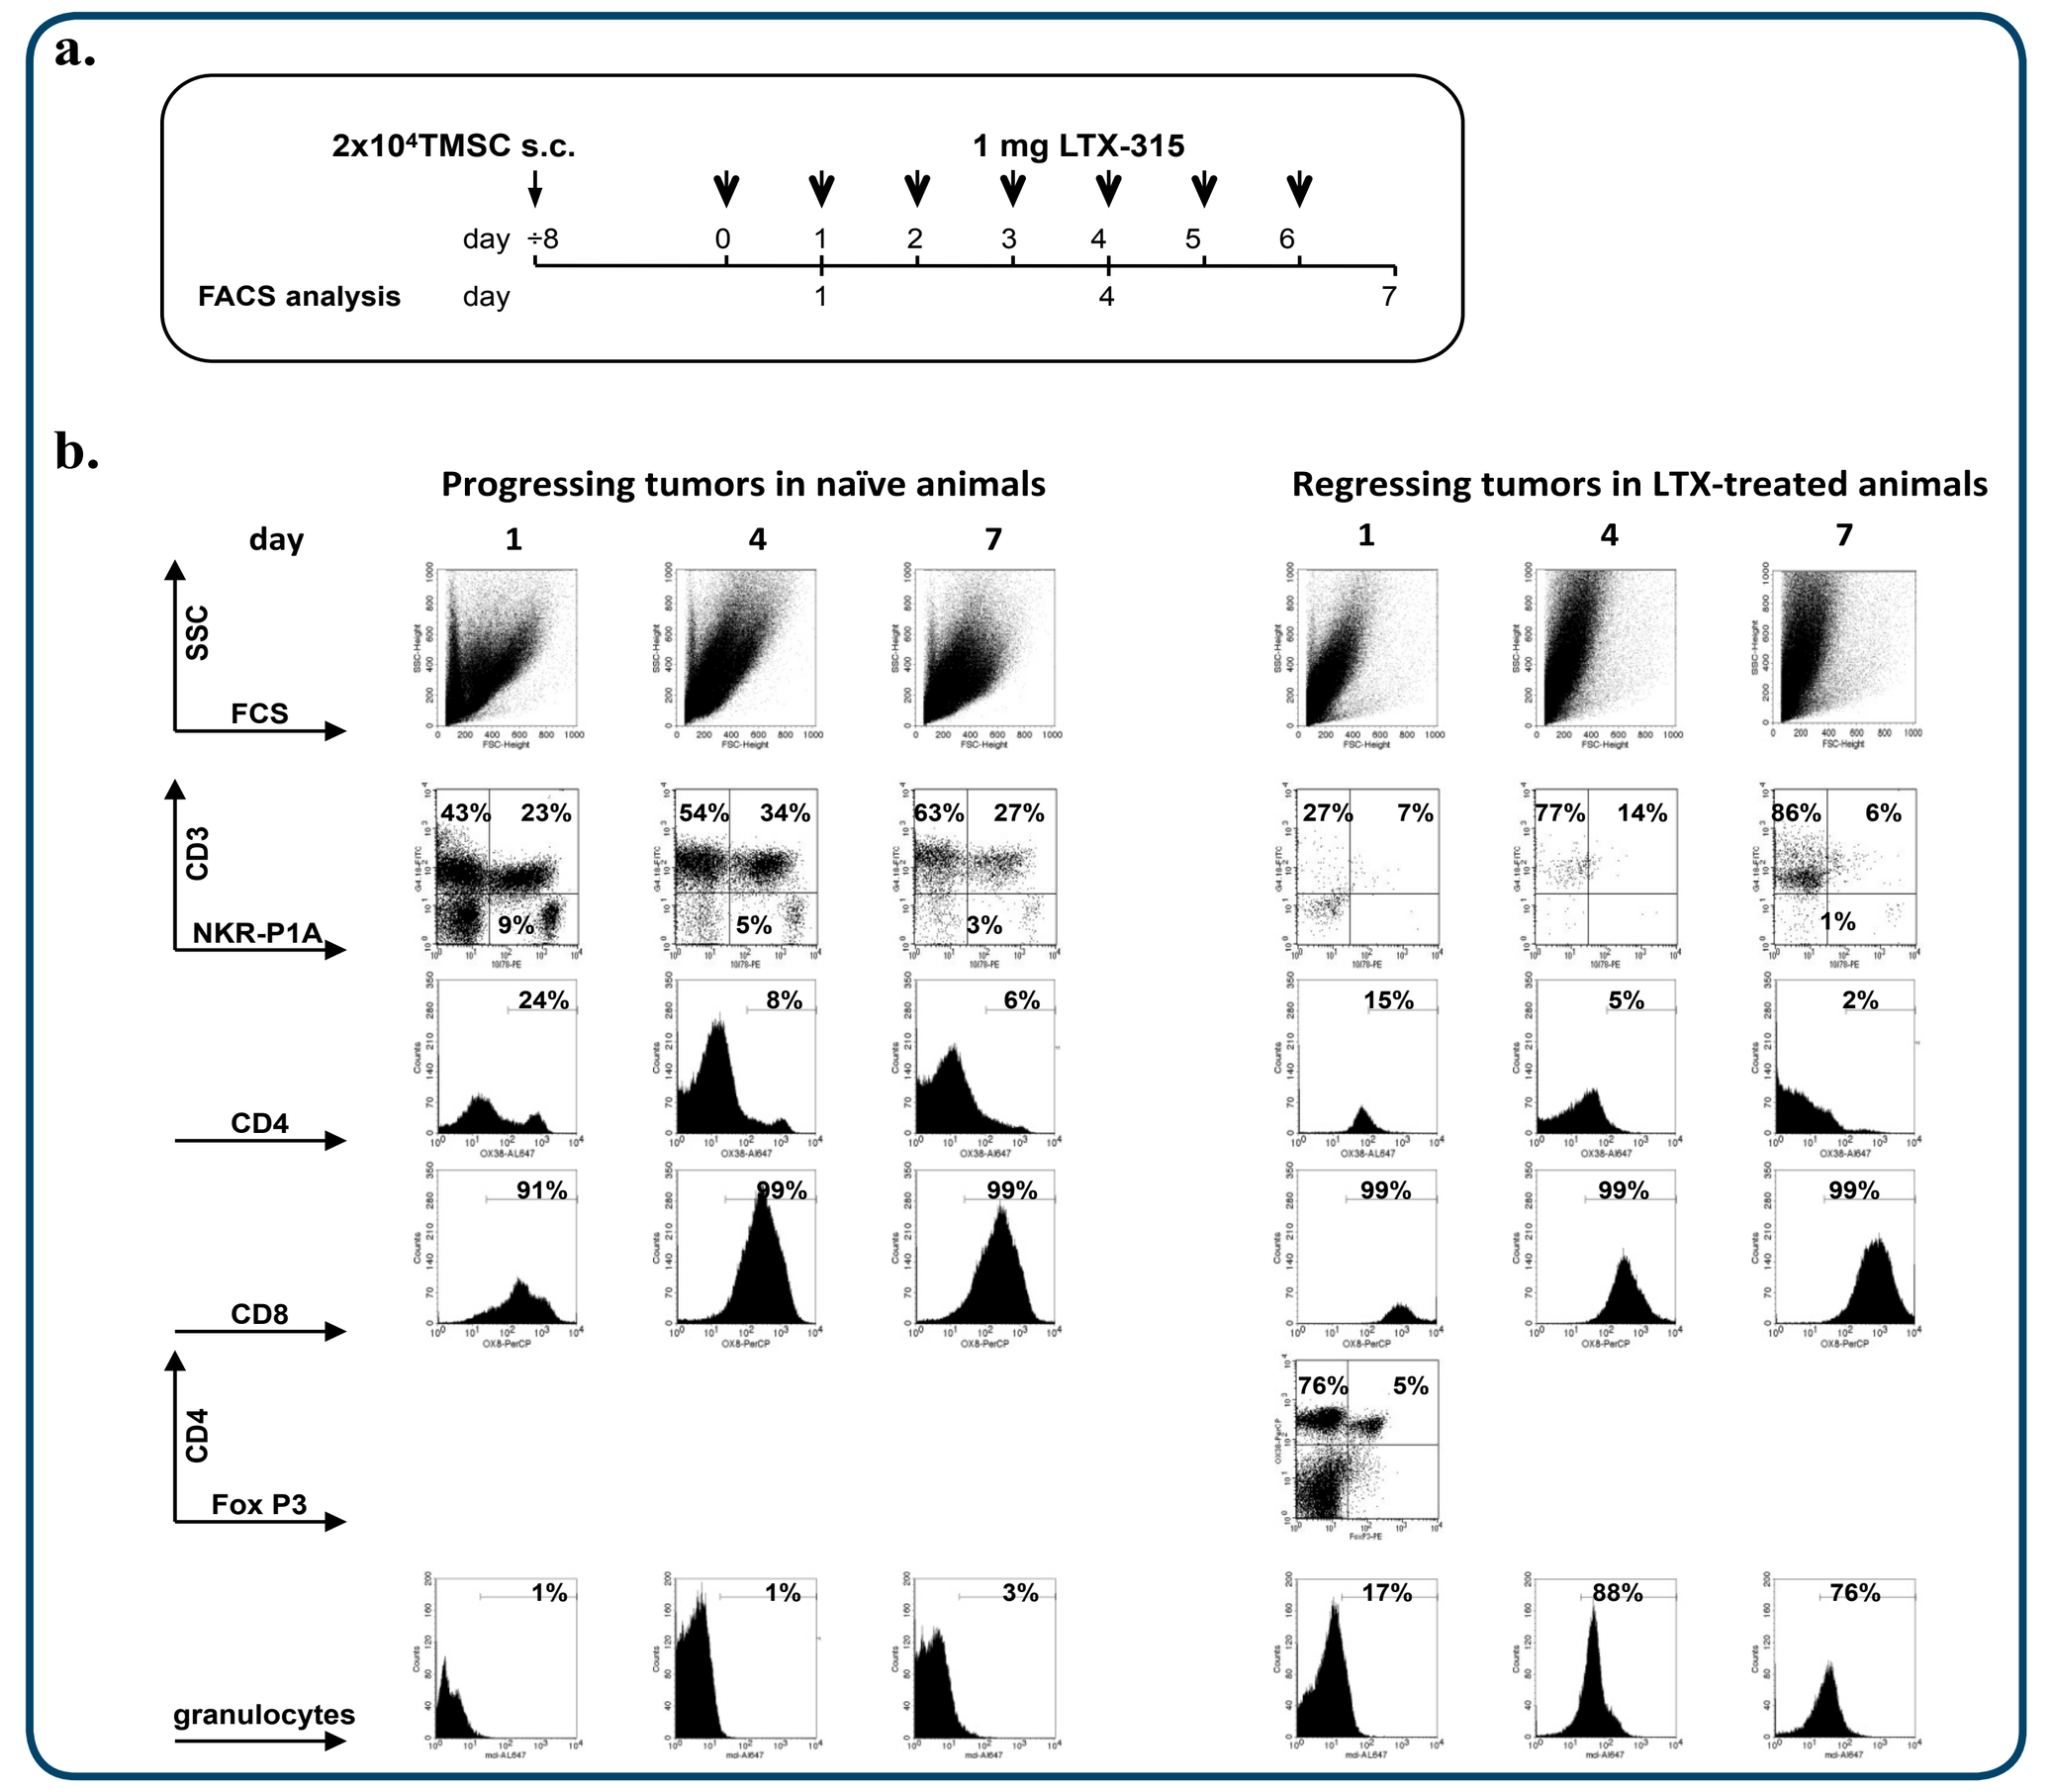
2x10⁴TMSC s.c.
1 mg LTX-315
FACS analysis
Progressing tumors in naïve animals
Regressing tumors in LTX-treated animals
SSC
FCS
CD3
NKR-P1A
CD4
CD8
Fox P3
granulocytes

Animals were inoculated with $2{\cdot}10^6$ TMSC cells s.c. Eight days later, the animals were given i.t. injections of 1 mg LTX-315 daily for 7 days. Control group received saline. Tumors were resected at indicated days and processed for flow cytometric analysis.

Figure 6. LTX-315 treatment results in the release of cytokines involved in inflammatory responses

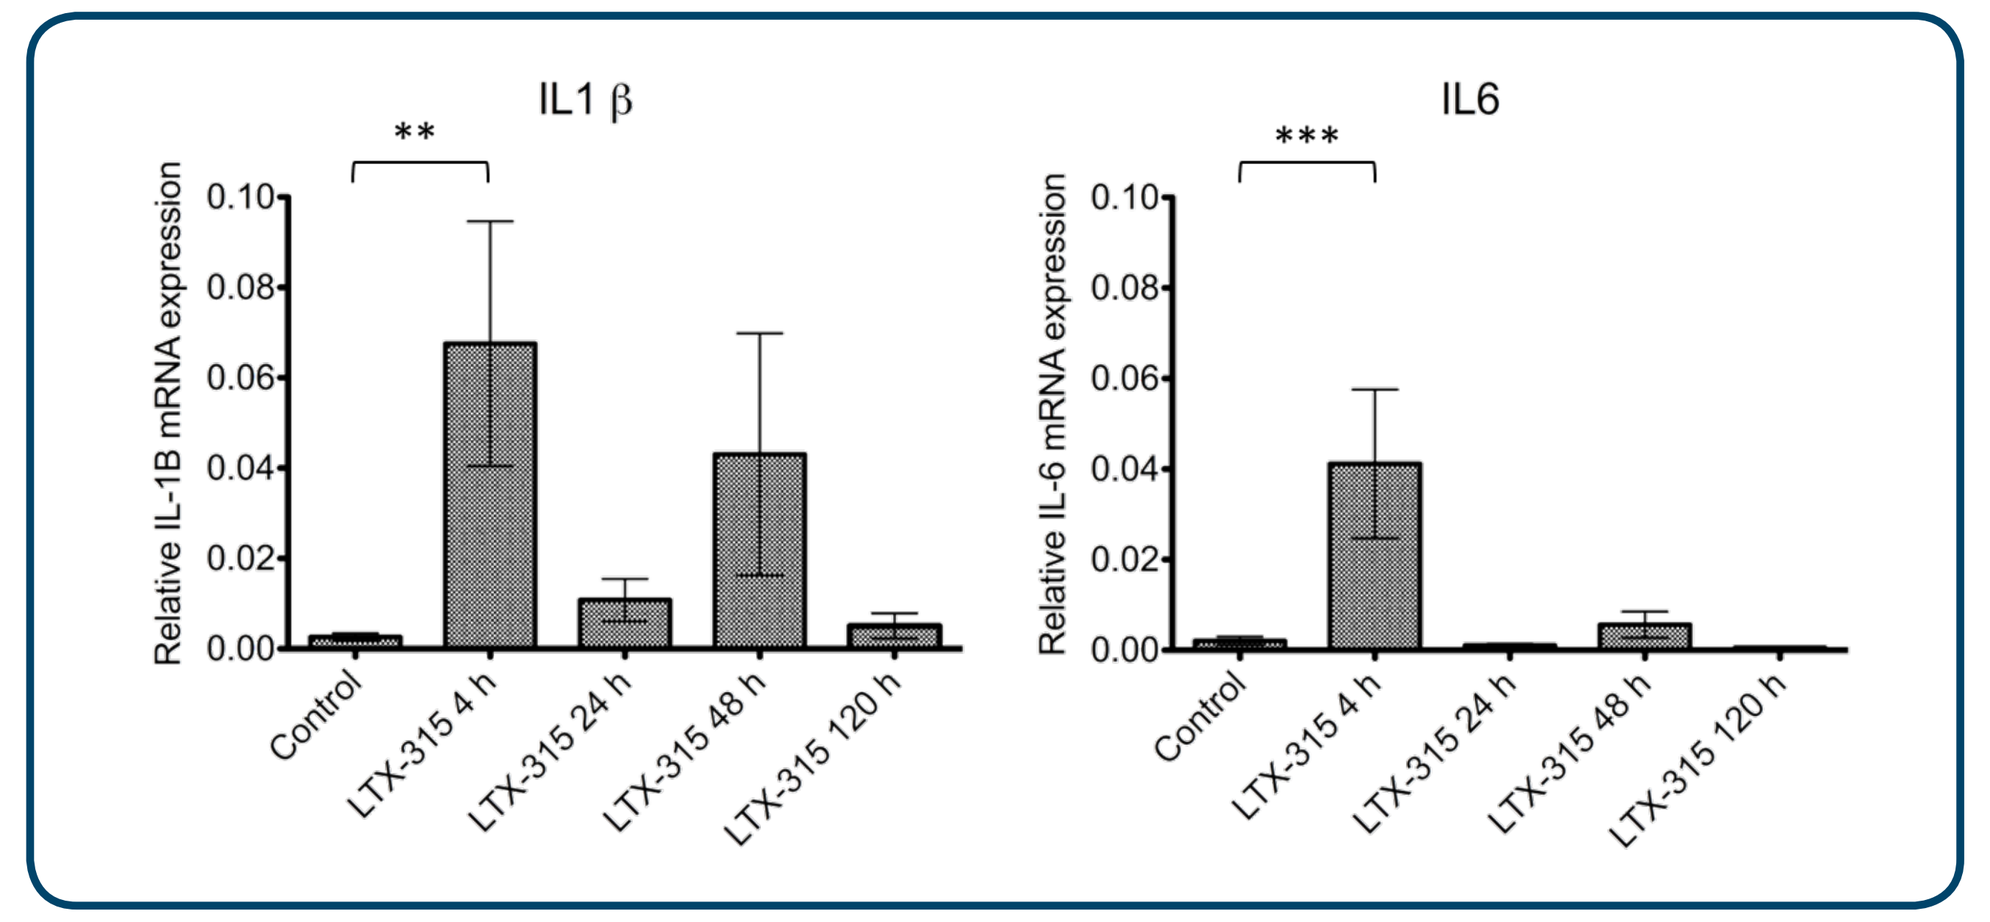
IL1 β
IL6
Relative IL-1B mRNA expression
Relative IL-6 mRNA expression
Control
LTX-315 4 h
LTX-315 24 h
LTX-315 48 h
LTX-315 120 h

- Following a single intratumoral injection of saline or 1mg LTX-315 in a B16F1 melanoma, tumor tissue were harvested at different time points and analyzed for mRNA expression of different cytokines using real time PCR. Data from three animals are presented for each time point as mean ±

Figure 7. Transfer of immunity by splenocytes from animals cured by LTX-315

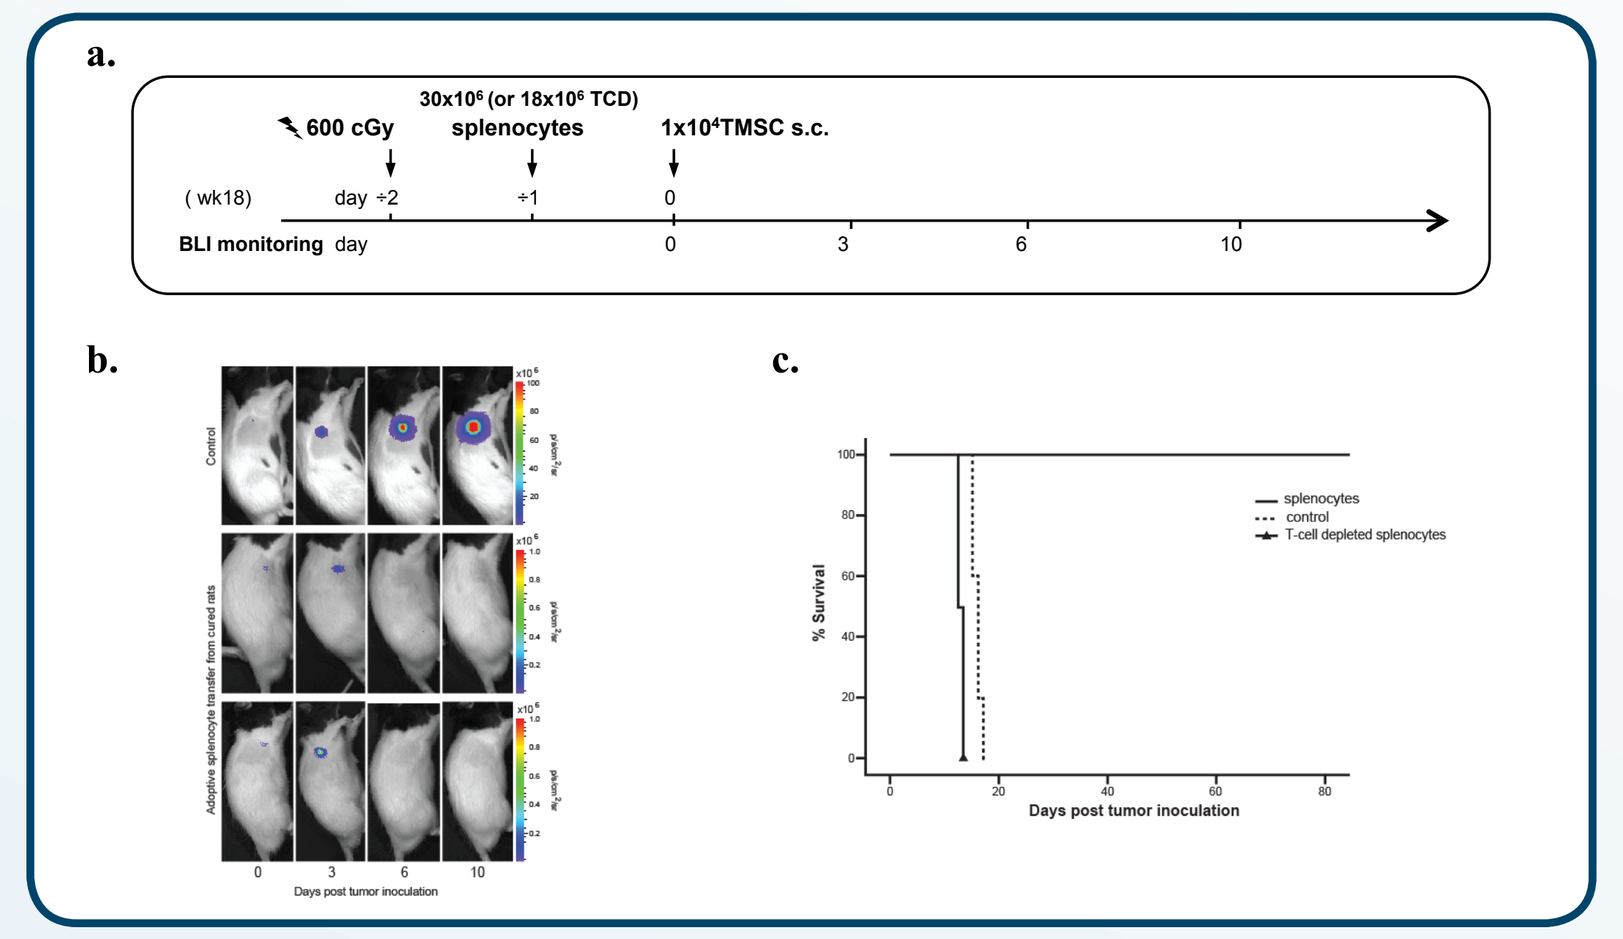
600 cGy
30x10⁶ (or 18x10⁶ TCD) splenocytes
1x10⁴TMSC s.c.
BLI monitoring
% Survival
Days post tumor inoculation

a) Rats received either isolated spleen cells from cured rats (n=15), or T-cell-depleted spleen cells (n=3). Control animals received spleen cells from naïve rats (n=5). One day after injection of spleen cells, animals were inoculated with TMSC s.c.

b) Representative images of tumor growth.

c) Survival curves

Fig 8. Protective immune responses in a rat hepatocellular carcinoma model

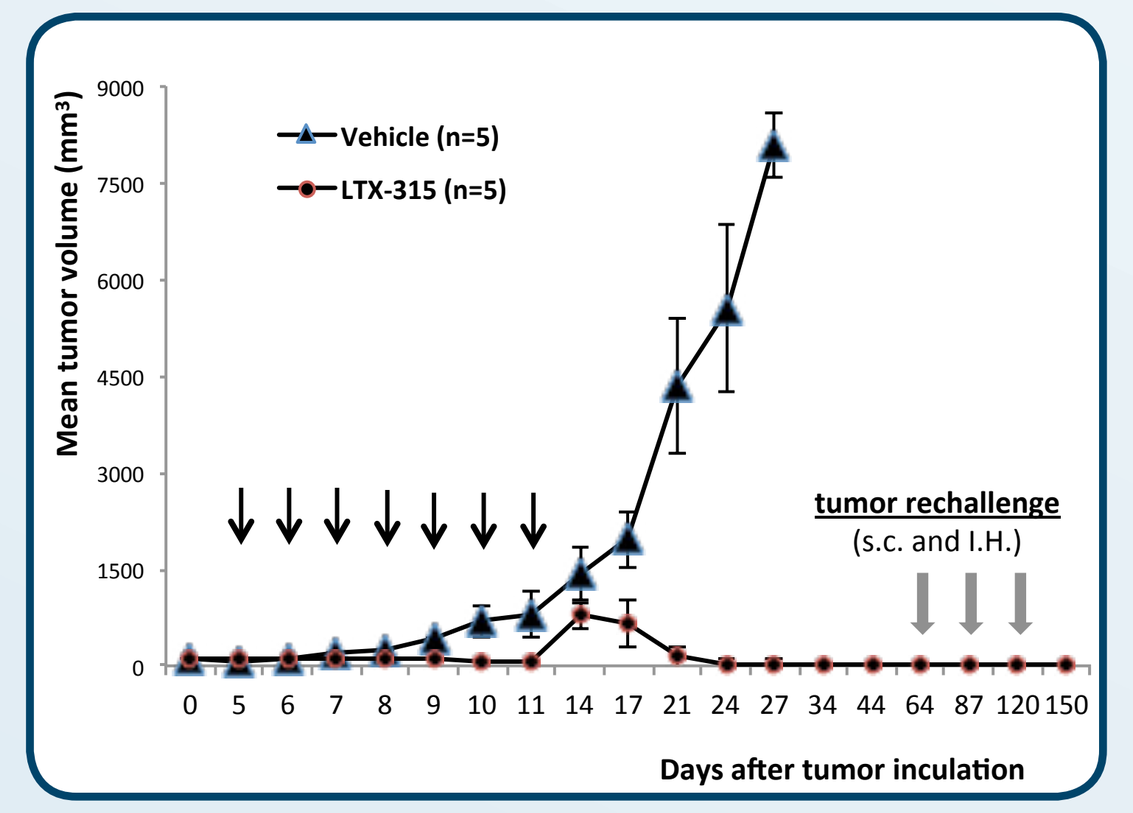
Vehicle (n=5)
LTX-315 (n=5)
Mean tumor volume (mm³)
tumor rechallenge
(s.c. and I.H.)
Days after tumor inculation

- HCC tumors were established by s.c. inoculation of HCC cells (JM1) in the flank of syngeneic Fischer 344 rats.
- The rats (n=5) were treated i.t. with 1-1.5 mg LTX-315 for 7 consecutive days. Control rats (n=5) were treated with 50 µL saline.
- Four weeks after complete tumor regression, the animals were re-chal¬lenged with HCC cells (JM1), both s.c. and intrahepatic.

Fig 9. The effect of LTX-315 treatment combined with surgery a rat hepatocellular carcinoma model.

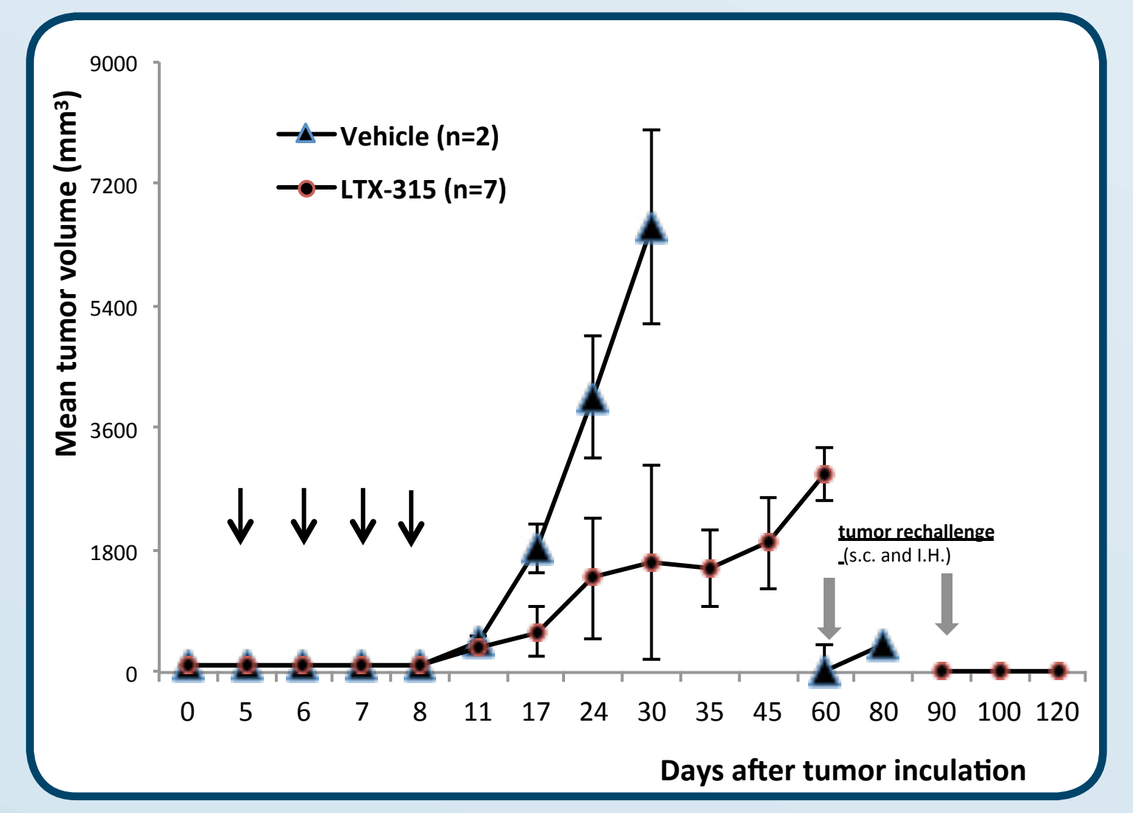
Vehicle (n=2)
LTX-315 (n=7)
Mean tumor volume (mm³)
tumor rechallenge
(s.c. and I.H.)
Days after tumor inculation

HCC tumors were established by s.c. inoculation of HCC cells (JM1) in syngeneic Fischer 344 rats.

The rats (n=7) were treated i.t. with 1mg LTX-315 for 4 consecutive days. Control rats (n=2) were treated with 50 µL saline. Surgical removal of tumor was performed with control rats at Day 30 and treated rats at Day 60.

Four weeks after complete tumor removal, the animals were re-challenged with HCC cells (JM1), both s.c. and intrahepatic.

Table 2. Effect of LTX-315 in rodent tumor models

Species	Tumor type	Number Inject.	Dose	Necrosis	Complete Regression	Immuno-protection
Syngenic Mice	A20 B-lymphoma	3	0,5-2 mg	Yes	Yes	Yes
	B16 melanoma	3	1 mg	Yes	Yes	Yes
	CT 26 Colon Carcinoma	3	0,5 – 2mg	Yes	Yes	Yes
Human Xenograft Nude mice	MCF-7 Breast Carcinoma	3	0,75 – 1mg	Yes	Yes	-
	SK MEL-5 Melanoma	3	0,75 – 1mg	Yes	No	-
Syngenic rats	trMSC, Transformed mesenchymal sarcoma	4-6	1-2 mg	Yes	Yes	Yes
	Hepatocellular Carcinoma	6-8	1 mg	Yes	Yes	Yes

Conclusion

- LTX-315 may represent a new potential drug for treatment of chemo-resistant solid tumors.
- Systemic protective and lasting immune responses have been obtained in several types of experimental tumor models. Furthermore, the adoptive transfer studies in the rTMSC model strongly implicates that T cells are instrumental in the systemic immune protection.
- Protective immune responses could also be achieved in partial responders in the HCC model indicating that LTX-315 has a potential as a neo-adjuvant combined with surgery and other treatment modalities for liver cancer.
- A phase 1 study has been completed with LTX-315 (data under analysis) and a new clinical study is in progress.
- LTX-315 may represents a novel strategy for personalized in situ vaccination against cancer.

References

1. Utsugi et al., Cancer Res. 1991, 1;51(11):3062-6
2. Woehlecke et al., Biochem J. 2003, 1;376(Pt 2):489-95
3. Kirszberg et al., Melanoma Res. 2009 Oct;19(5):301-8.
4. Cichorek et al., Folia Histochem Cytobiol. 2000;38(1):41-3.
5. Schröder-Borm et al., FEBS Lett. 2005 Nov 7;579(27):6128-34.
6. Fadnes et al., Glycoconj J. 2012 Oct;29(7):513-23.
7. Burdick et al., J Biol Chem. 1997 Sep 26;272(39):24198-202

Lytix Biopharma AS | P.O. Box 6447 | NO-9294 Tromsø, Norway | E-mail: oystein.rekdal@lytixbiopharma.com | Phone: +47 77 67 55 00 | Fax: +47 77 67 55 01

© WWW.CHILIGROUP.NO|2013